Molecular Shielding of the Pan-Hematopoietic Marker CD45 May Enable a Targeted Universal Approach for Replacement of the Hematopoietic System

Simon Garaudé[1], Romina Marone[1], Rosalba Lepore[1,2], Anna Devaux[1], Anna Camus[2], Izabela Durzynska[2,3], Eva-Maria Grossjohann[2], Andreja Knezevic[2], Ian Kirby[4], Patrick H. Van Berkel[4], Christian Kunz[3], Stefanie Urlinger[2] and Lukas T. Jeker[1]

[1]University of Basel and Basel University Hospital, Department of Biomedicine, Basel, Switzerland, [2]Cimeio Therapeutics, Basel, Switzerland, [3]Ridgeline Discovery GmbH, Basel, Switzerland, [4]ADC Therapeutics (UK) Ltd, London, UK

Introduction

- The pan-hematopoietic marker CD45 is critical for the function of immune cells (i.e, CD45 mutations can lead to severe combined immunodeficiency (SCID)).
- CD45-targeted surrogate antibody-drug conjugates (ADCs) enable chemotherapy-free hematopoietic stem cell transplantation (HSCT) in mice[1].
- Complete eradication of CD45+ cells could represent a universal approach to reset the hematopoietic system for hematologic malignancies or severe autoimmune diseases - independent of the cellular origin of a hematopoietic disease.
- CD45-targeted cell depleting therapies are currently limited to a single pre HSCT dose since continued full depletion would be detrimental. However, targeted post HSCT therapy would be desirable for disease control.
- Here, we describe the identification of a CD45 base editable shielding variant paired with the development of a new and potent CD45-targeting ADC (CIM053-ADC). When engineered into HSPCs this CD45 variant may enable safe post HSCT CD45-targeted therapy.

Results

1. Identification of base editable regions in CD45 extracellular domains that could provide shielding from targeted therapies

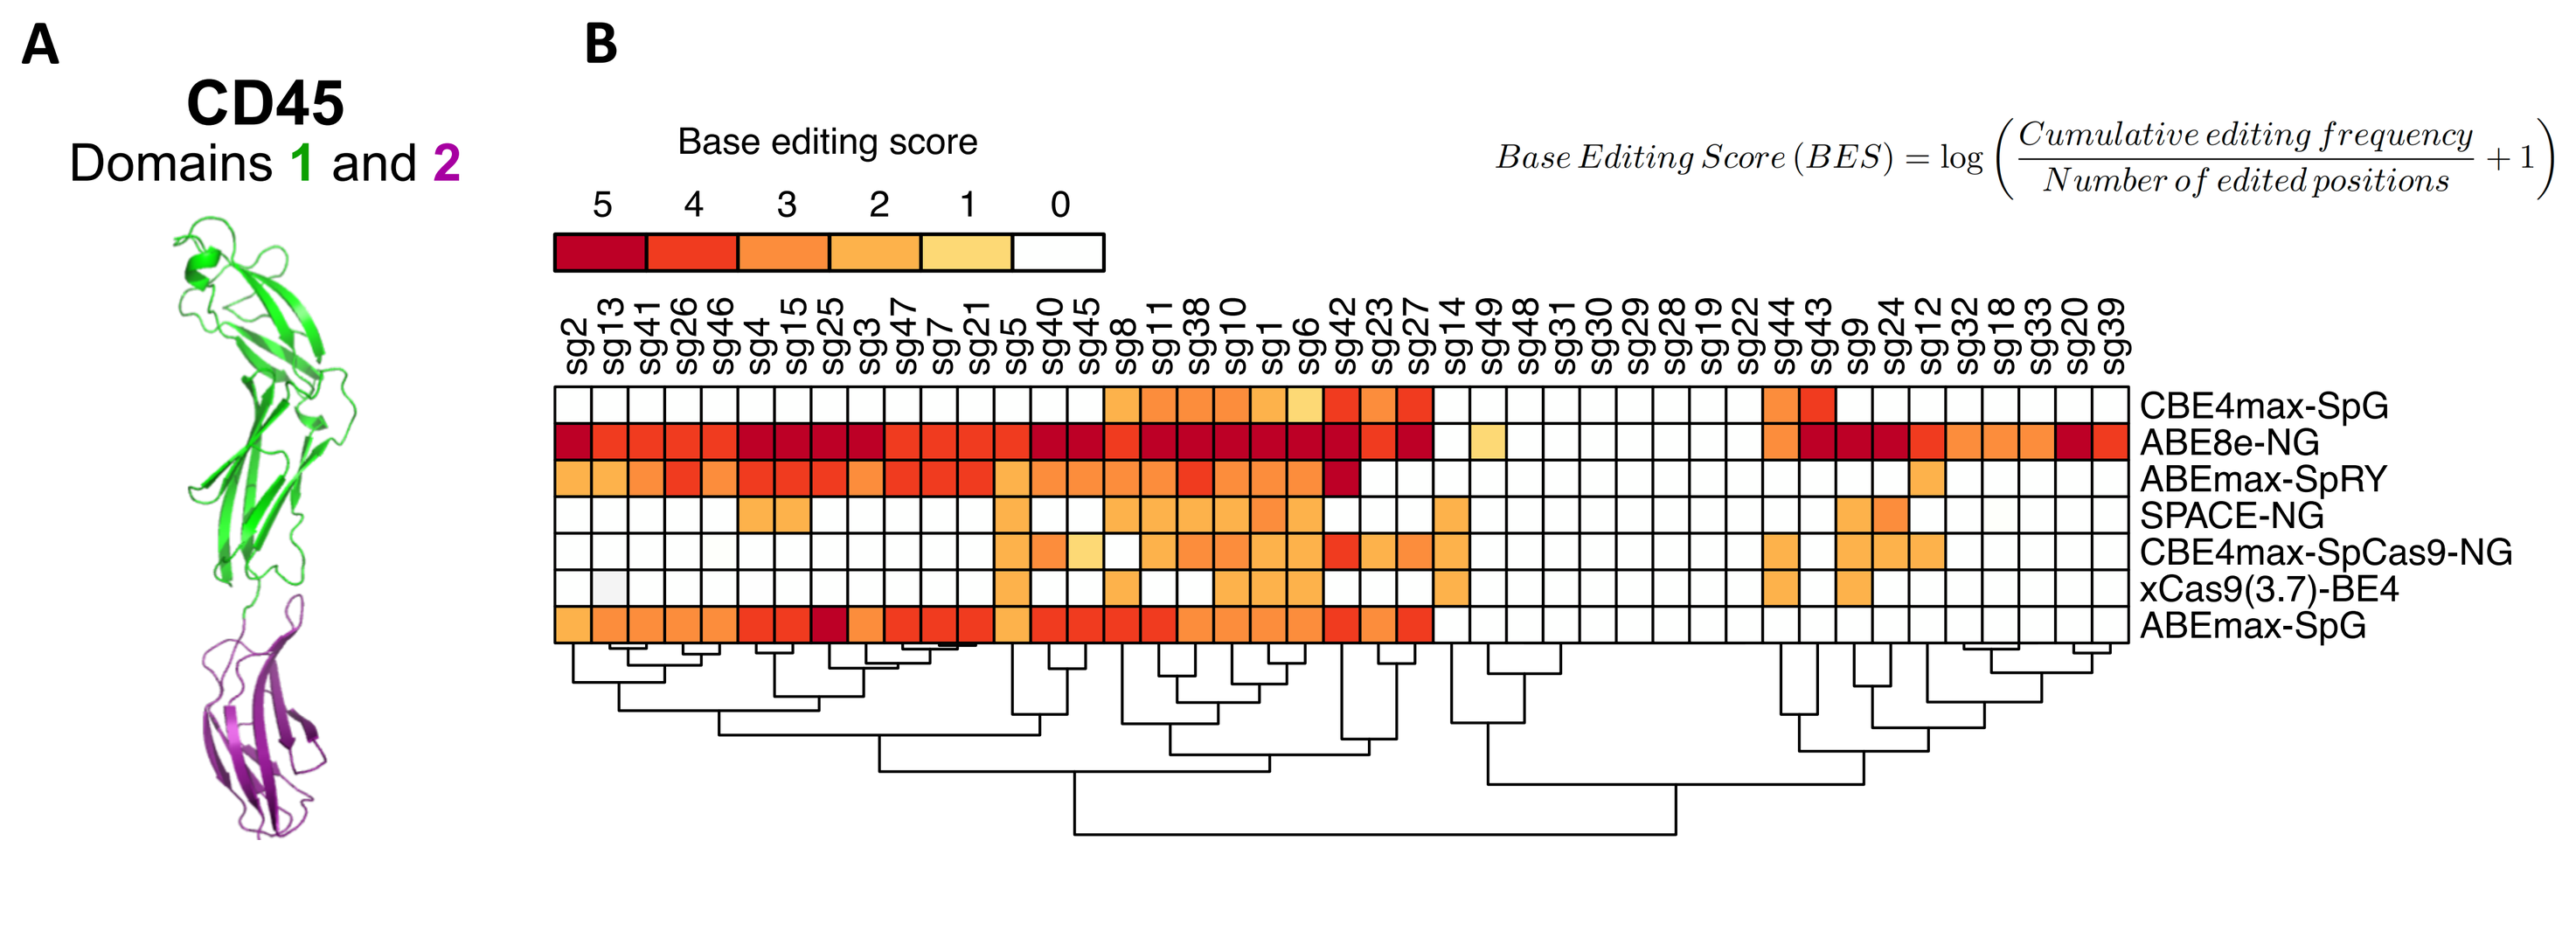
A
CD45
Domains 1 and 2
B
Base editing score
5 4 3 2 1 0
$Base\ Editing\ Score\ (BES) = \log\left(\frac{Cumulative\ editing\ frequency}{Number\ of\ edited\ positions} + 1\right)$
CBE4max-SpG
ABE8e-NG
ABEmax-SpRY
SPACE-NG
CBE4max-SpCas9-NG
xCas9(3.7)-BE4
ABEmax-SpG

A: Crystal structure of CD45 domains 1+2 (PDB: 5FMV); CD45 domains 1+2 were computationally identified as suitable for targeted therapy and contain antibody A and B epitopes **B:** Base editor screening targeting computationally selected extracellular regions of CD45 domains 1+2 predicted to be amenable for shielding. (Plasmid-based screen in K562 cells; Sanger sequencing 3 days post-electroporation).

2. Variants 1 and 2 shield human T cells from antibody-drug-conjugate (ADC)

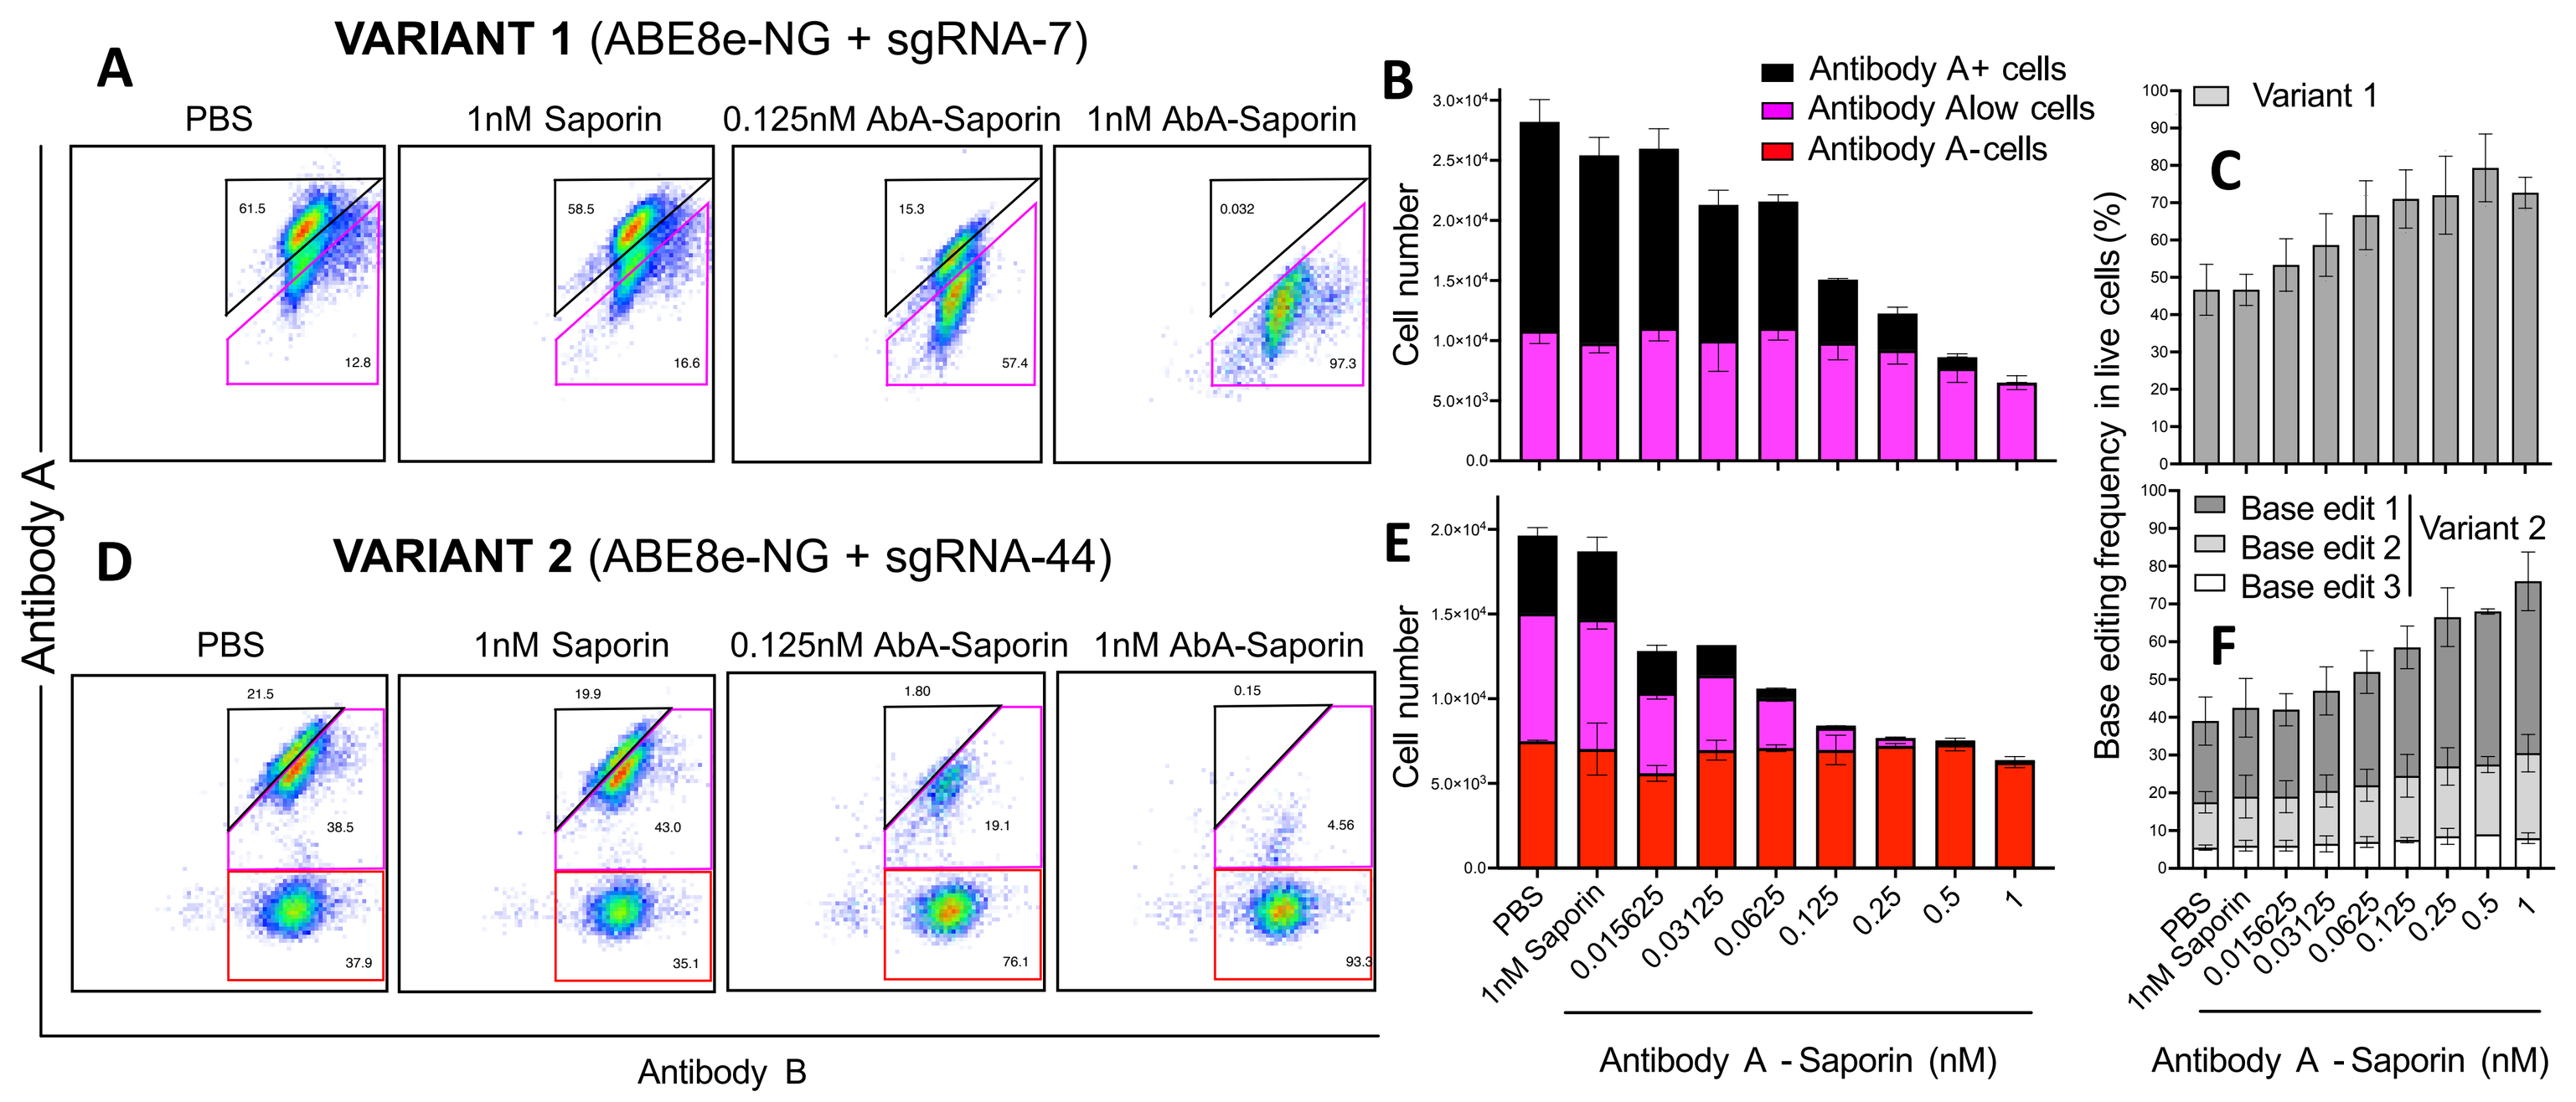
A
VARIANT 1 (ABE8e-NG + sgRNA-7)
PBS
1nM Saporin
0.125nM AbA-Saporin
1nM AbA-Saporin
D
VARIANT 2 (ABE8e-NG + sgRNA-44)
Antibody A
Antibody B
B
E
Antibody A+ cells
Antibody Alow cells
Antibody A-cells
Cell number
Antibody A - Saporin (nM)
C
F
Variant 1
Base edit 1
Base edit 2
Base edit 3
Variant 2
Base editing frequency in live cells (%)

A/D: Flow cytometry profiles of variant 1 and 2 bulk base edited T cells after 72h of incubation with antibody A-Saporin (ADC) **B/E:** Quantification of A and B. **C/F:** Sanger sequencing of sorted live cells confirms enrichment of variant 1 and 2 base edits with increasing ADC concentrations

3. Variant 3 shows favorable biophysical properties and completely abolishes binding to antibody B

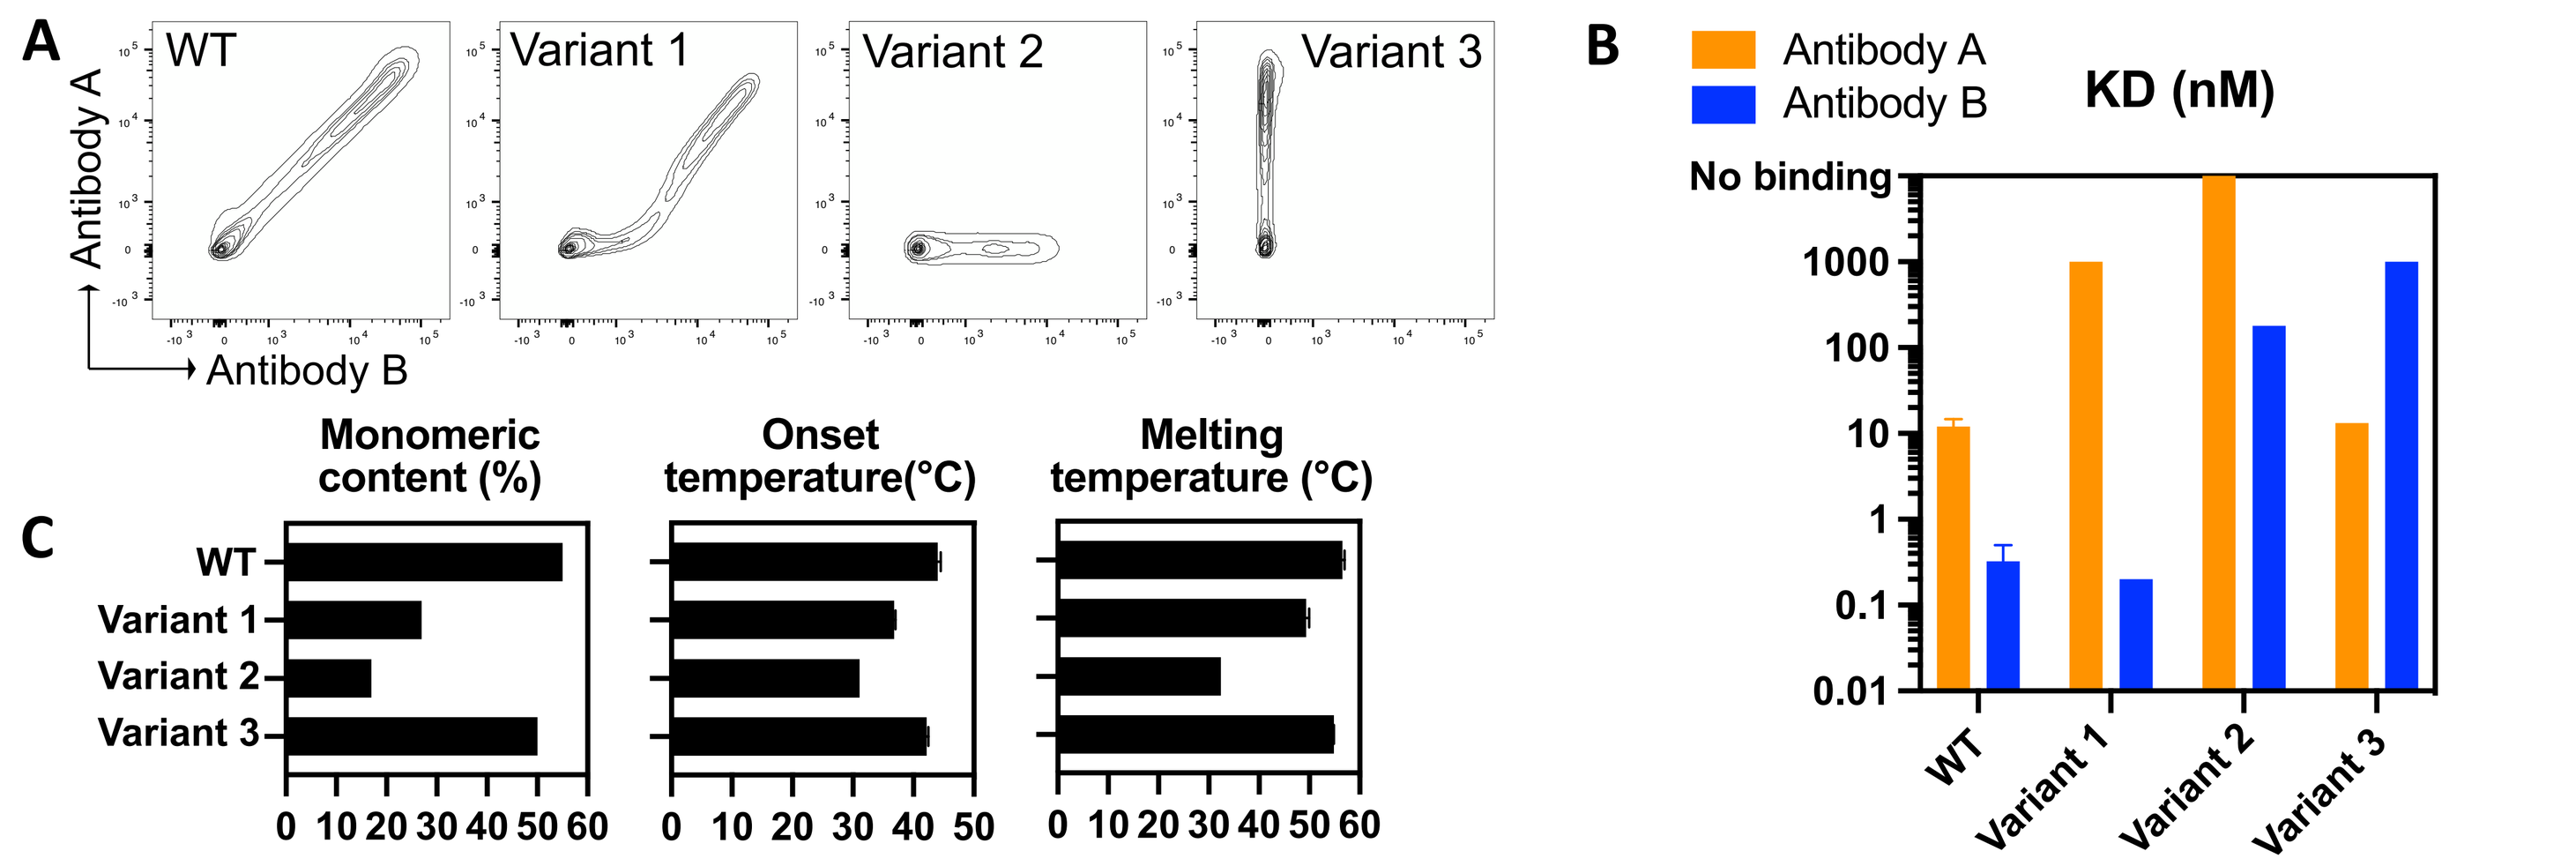
A
WT
Variant 1
Variant 2
Variant 3
Antibody A
Antibody B
B
Antibody A
Antibody B
KD (nM)
No binding
C
Monomeric content (%)
Onset temperature(°C)
Melting temperature (°C)
WT
Variant 1
Variant 2
Variant 3

A: Flow cytometry of DF-1 cells transiently overexpressing CD45 wildtype and variants confirms reduced/abolished antibody binding to CD45 variants **B:** Real-time affinity determinations of monovalent, biomolecular interactions using biolayer interferometry confirm 100- to >1000fold reduced binding affinity (KD). **C:** Biophysical protein characterization shows that Variant 3 retains protein stability and low aggregation propensity while Variants 1 and 2 have reduced protein integrity.

4. Base editing in CD34+ hematopoietic stem and progenitor cells (HSPCs) in vitro

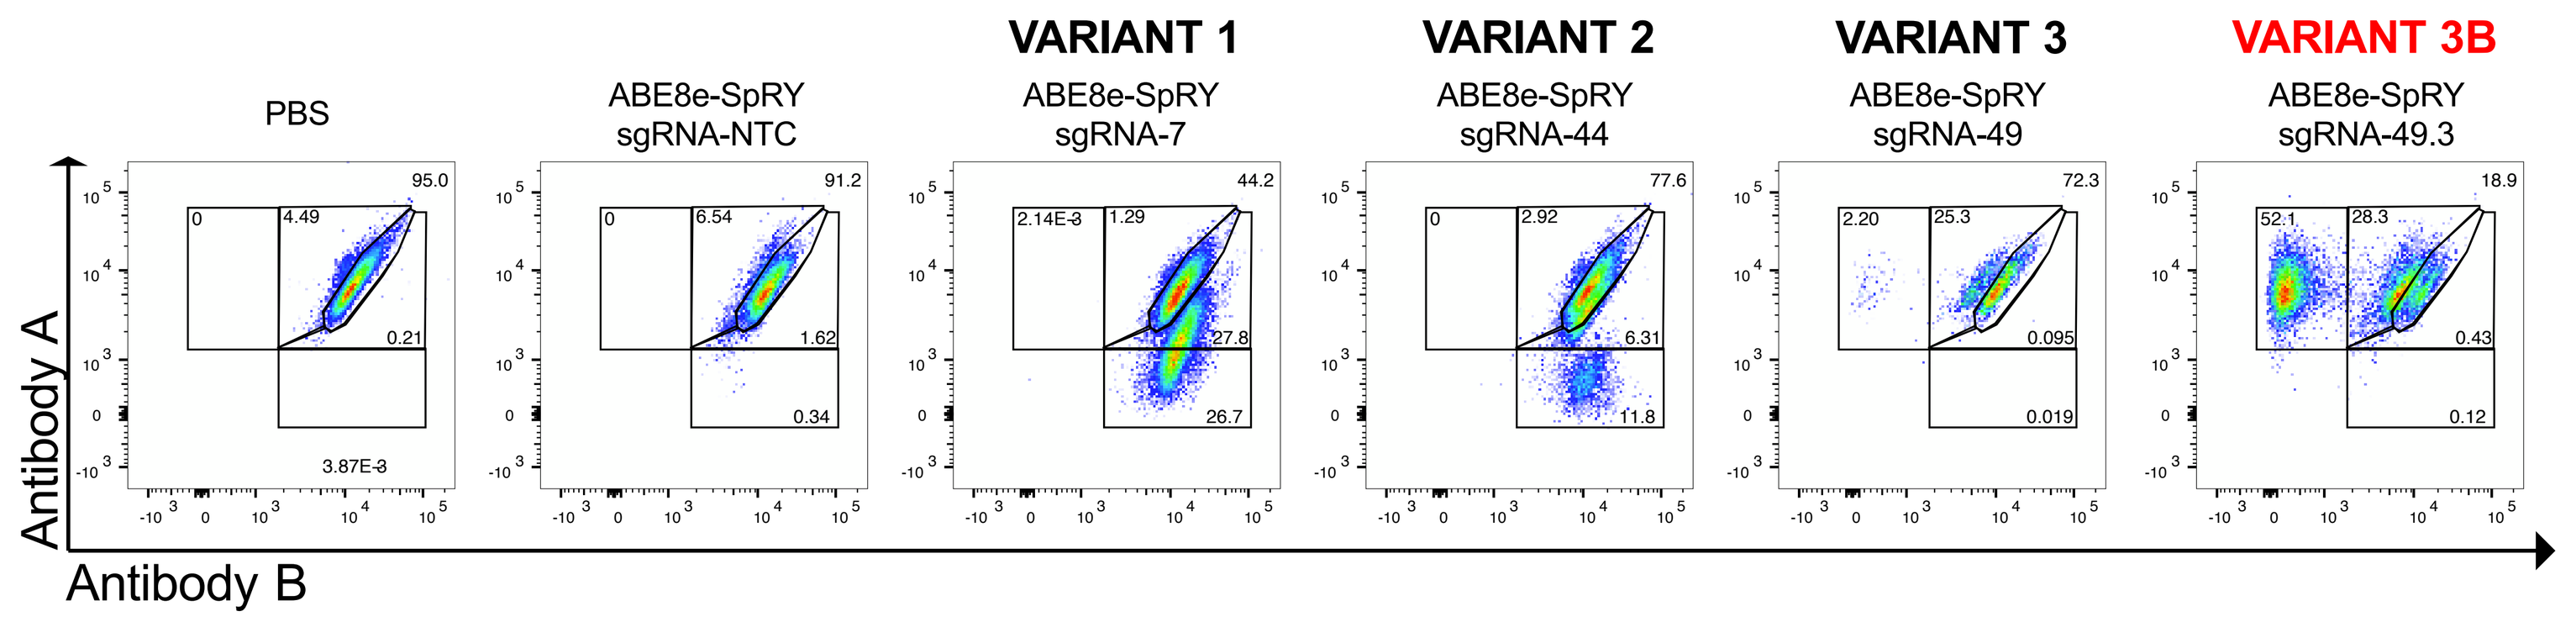
PBS
ABE8e-SpRY sgRNA-NTC
VARIANT 1
ABE8e-SpRY sgRNA-7
VARIANT 2
ABE8e-SpRY sgRNA-44
VARIANT 3
ABE8e-SpRY sgRNA-49
VARIANT 3B
ABE8e-SpRY sgRNA-49.3
Antibody A
Antibody B

Flow cytometry profile of base edited CD34+ HSPCs 12 days post-electroporation with base editor mRNA and sgRNA. Variant 3 editing efficiency was improved by repositioning sgRNAs using Cas9-SpRY's relaxed PAM recognition capabilities[2]. sgRNA-49.3 reduced bystander editing (not shown) and increased on-target editing (25-fold) resulting in Variant 3B.

5. Variant 3B shields HSPCs from CIM053-ADC killing while unedited HSPCs and co-cultured tumor cells are selectively depleted

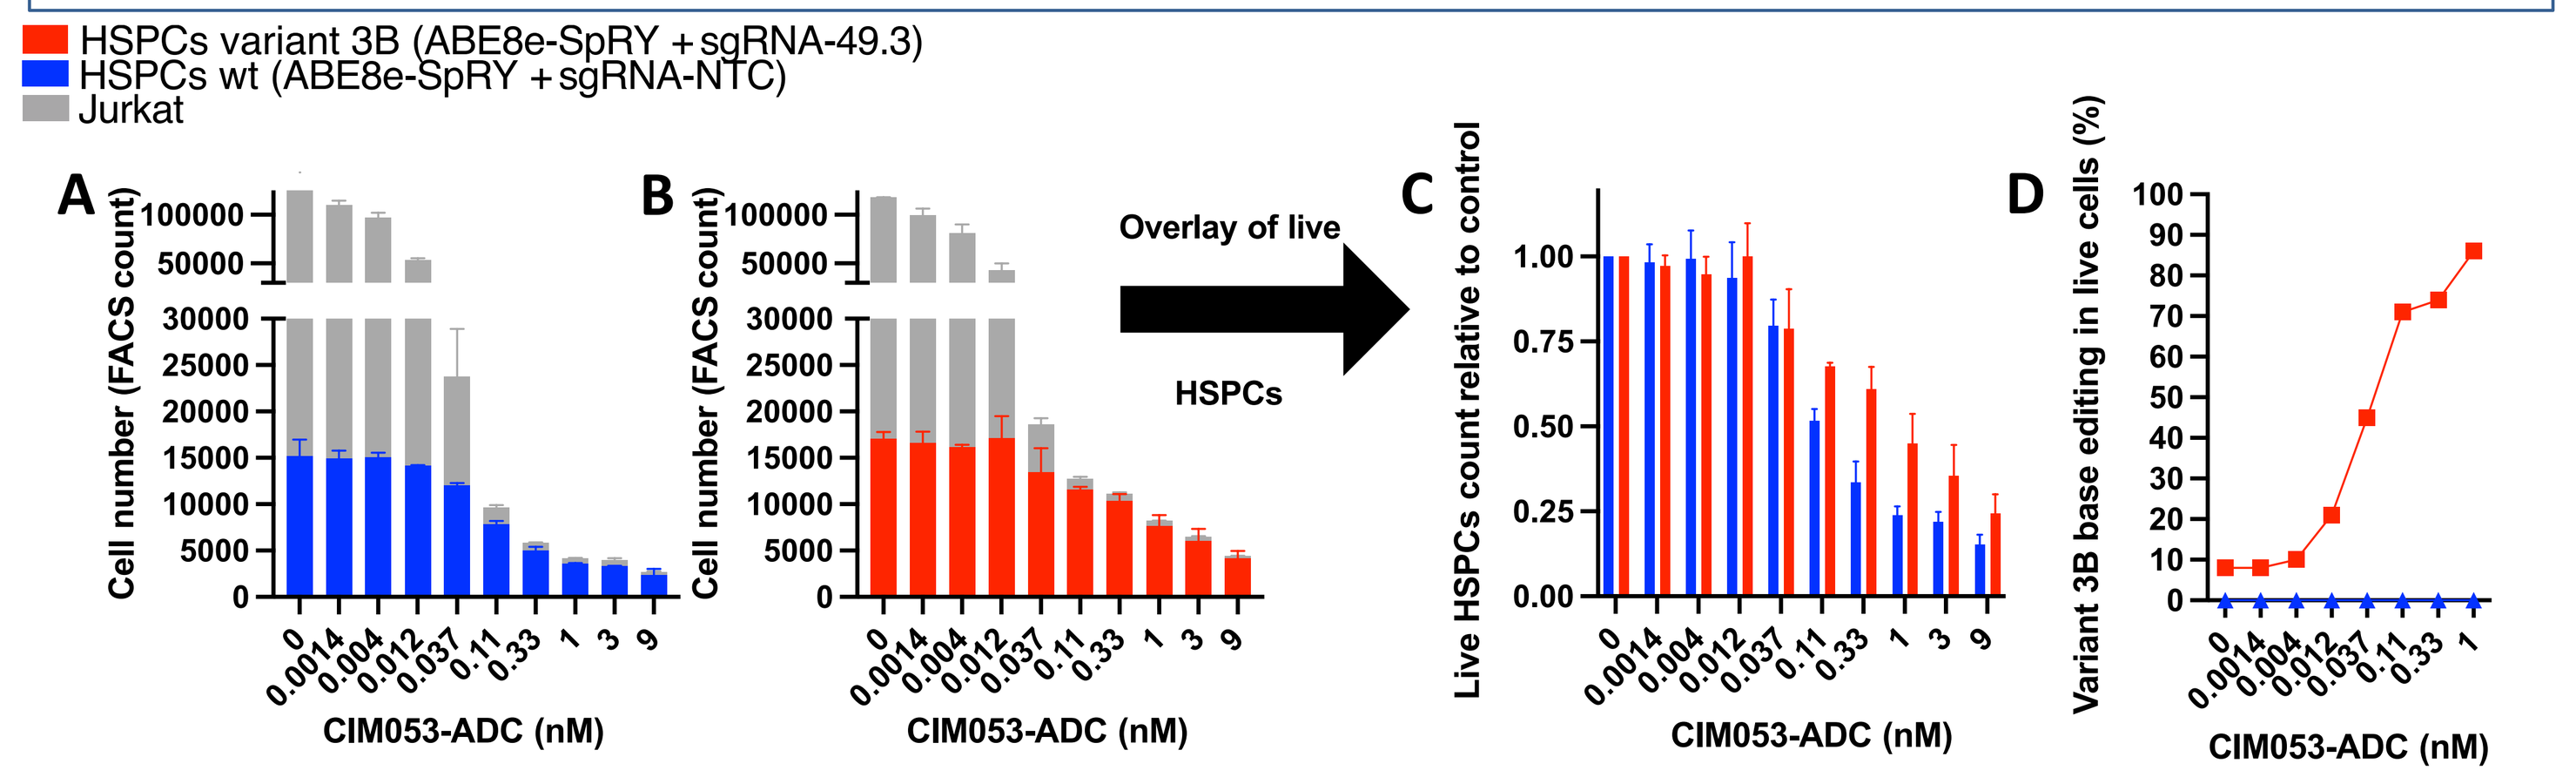
HSPCs variant 3B (ABE8e-SpRY + sgRNA-49.3)
HSPCs wt (ABE8e-SpRY + sgRNA-NTC)
Jurkat
A
B
C
D
Cell number (FACS count)
CIM053-ADC (nM)
Overlay of live
HSPCs
Live HSPCs count relative to control
Variant 3B base editing in live cells (%)

A/B: Absolute cell count of wt-HSPCs:Jurkat and Variant 3B-HSPCs:Jurkat co-cultures after 72h of incubation with CIM053-ADC concentrations, a novel humanized and engineered monoclonal antibody against hCD45 conjugated to a potent toxin (co-cultures seeded at 1:1 cell ratio) **C:** Comparison of HSPC survival ratios between A and B **D:** Sanger sequencing of all sorted live cells from co-culture conditions confirms enrichment of base-edited Variant 3B expressing HSPCs using CIM053-ADC at increasing concentrations.

6. Variant 3B shields hematopoietic cells from CIM053-ADC killing in vivo

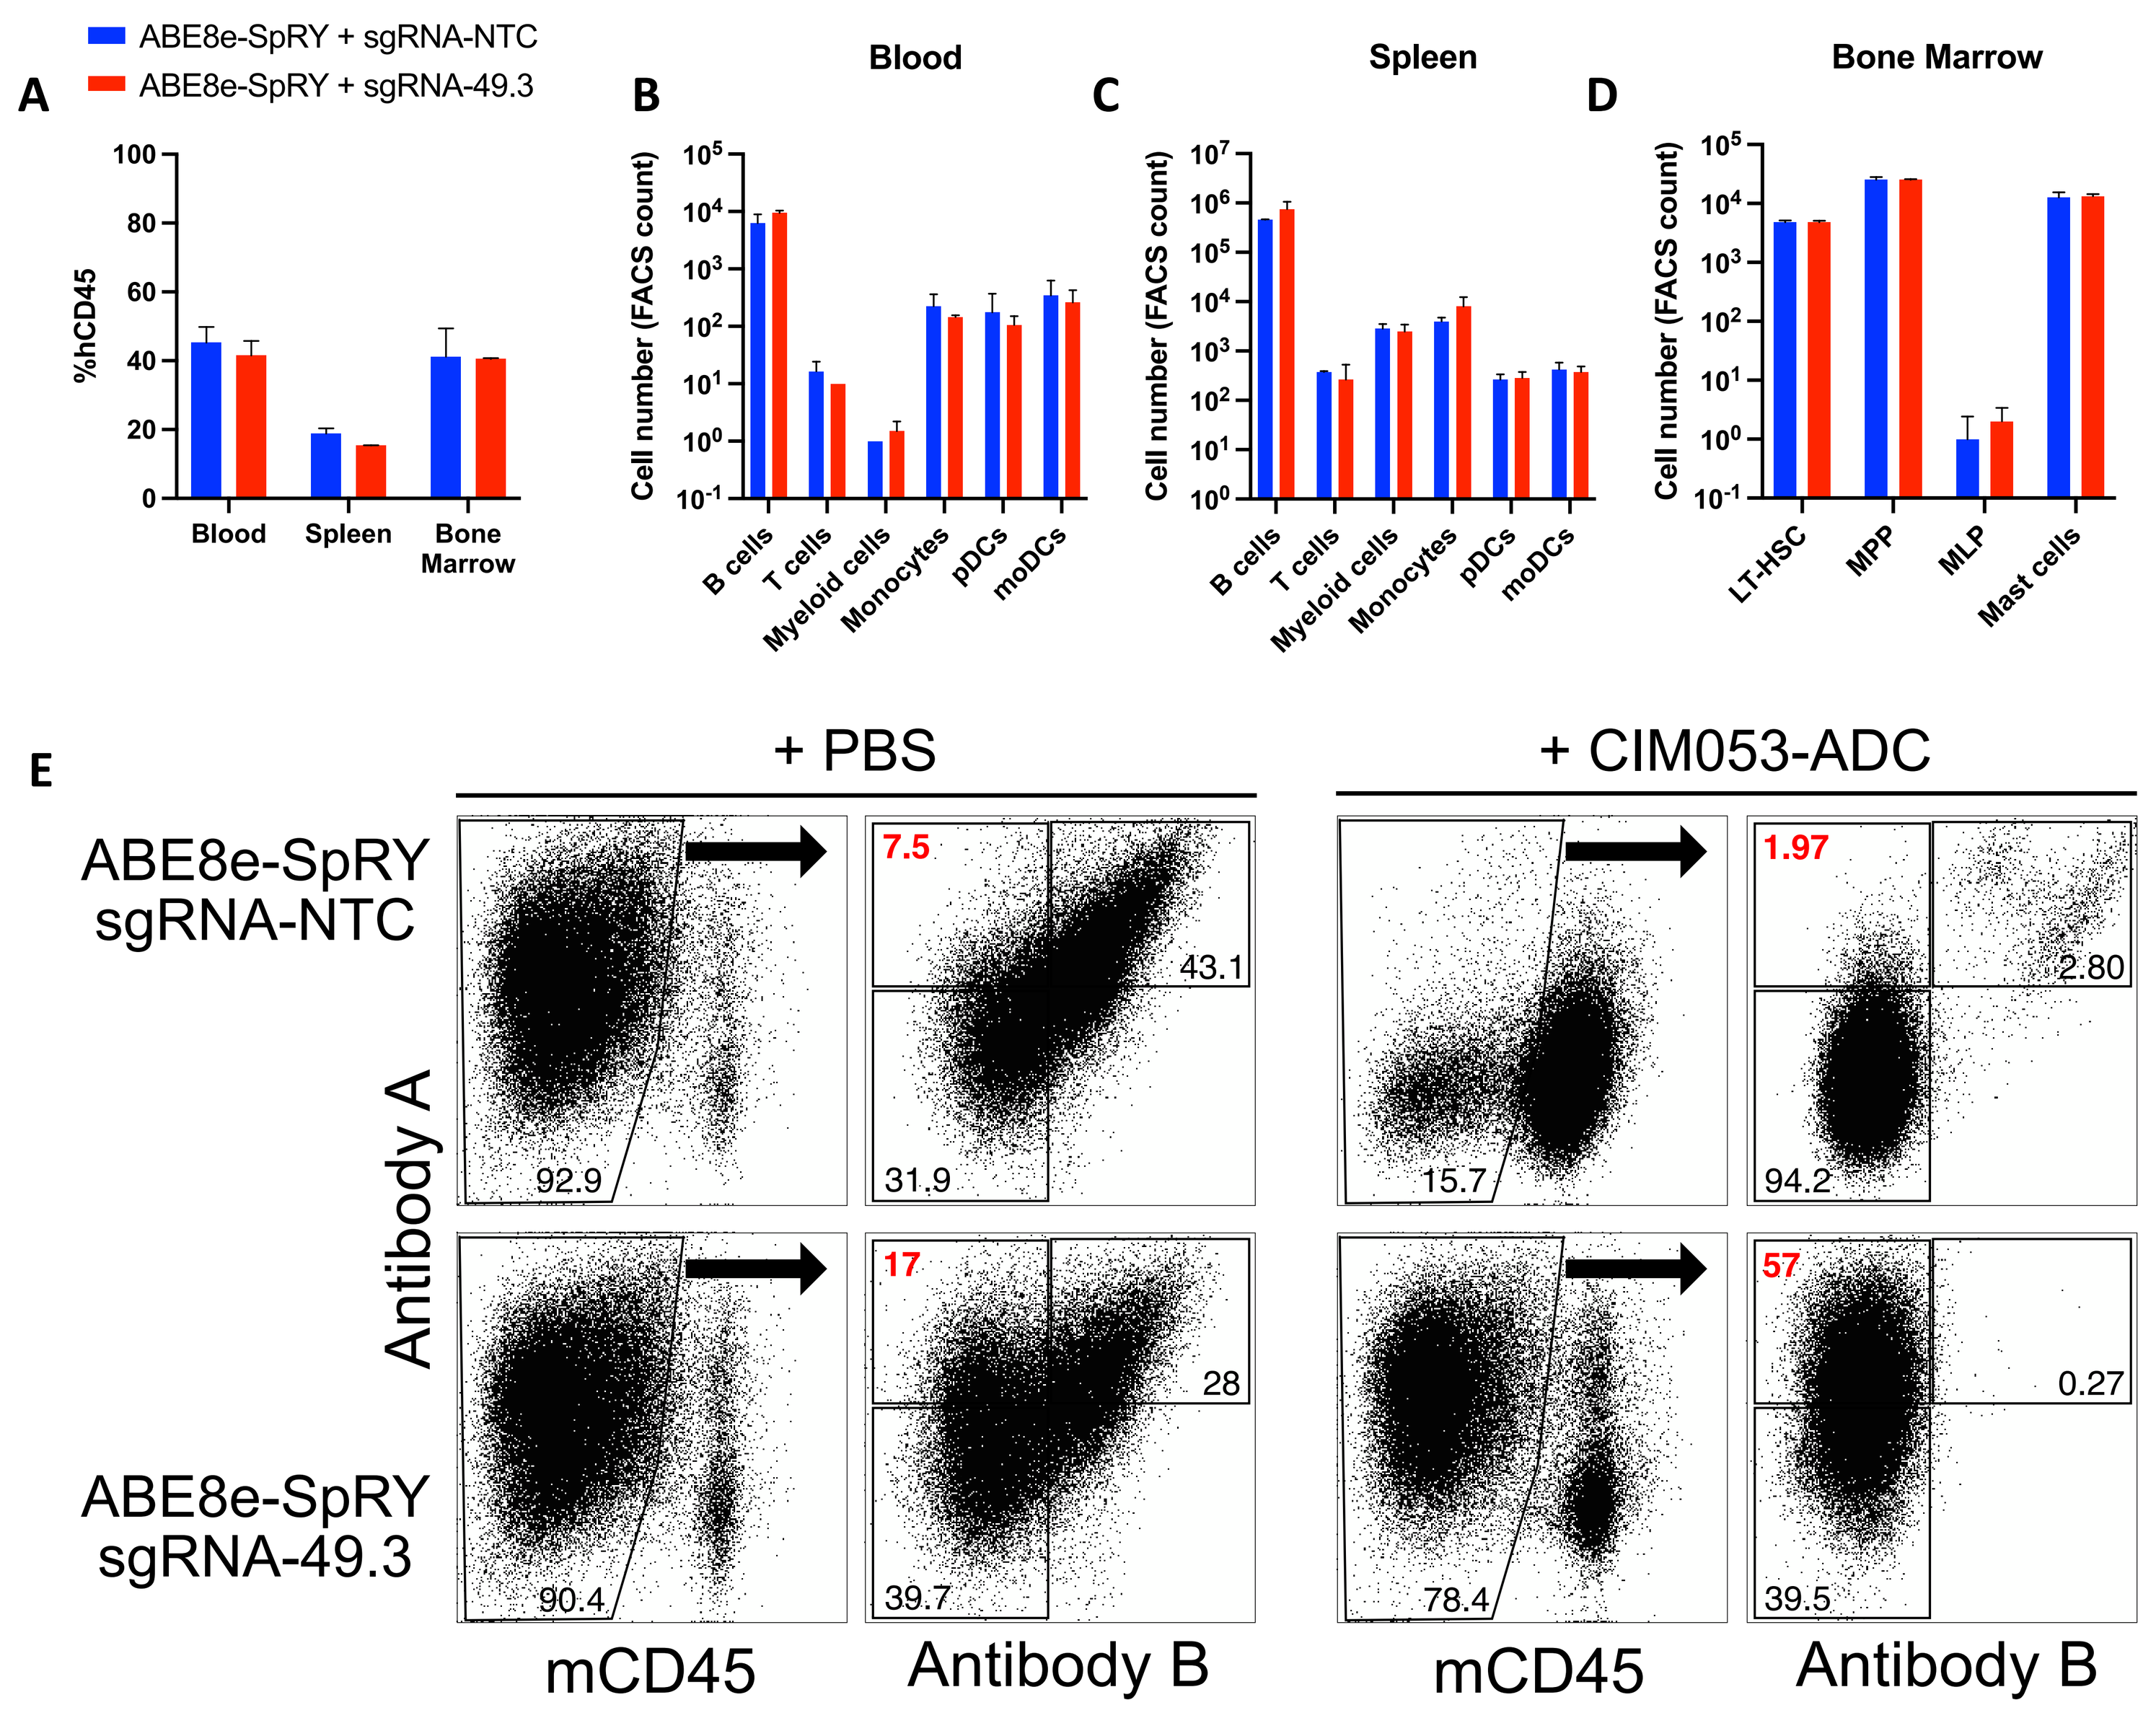
ABE8e-SpRY + sgRNA-NTC
ABE8e-SpRY + sgRNA-49.3
A
%hCD45
Blood
Spleen
Bone Marrow
B
Blood
C
Spleen
D
Bone Marrow
Cell number (FACS count)
B cells
T cells
Myeloid cells
Monocytes
pDCs
moDCs
LT-HSC
MPP
MLP
Mast cells
E
+ PBS
+ CIM053-ADC
ABE8e-SpRY sgRNA-NTC
ABE8e-SpRY sgRNA-49.3
Antibody A
mCD45
Antibody B

A: Human (antibody A)/mouse chimerism is comparable in mice reconstituted with wt-HSPCs or Variant 3B-HSPCs (12 weeks post HSPCs injection) demonstrating unaltered engraftment capacity of engineered HSPCs. **B/C/D:** Spleen, blood and bone marrow cell differentiation profiles are comparable between mice reconstituted with wt-HSPCs or Variant 3B-HSPCs **E:** Variant 3B shields bone marrow hematopoietic cells from CIM053-ADC killing in vivo.

Conclusions

- Using computational rational design in combination with guidance from alanine scanning as well as a comprehensive base editor screening, we identified amino acid substitutions that can be introduced by base editors to act as molecular shields. Edited cells (e.g. T cells, HSPCs) are resistant to targeting by CD45-directed ADCs in vitro.
- Biophysical characterization of shielding variants revealed that some amino acid substitutions impair the biophysical properties of CD45 and result in reduced/abolished binding of antibodies that are not directly interacting with the modified amino acid residue. This suggests that shielding might result from major conformational protein changes and not through the elimination of a direct interaction point.
- CD45 variant 3B completely shields from the novel humanized, engineered CIM053-ADC while maintaining intact protein properties.
- Co-culture of variant 3B edited HSPCs with tumor cells enables selective depletion of tumor and unedited HSPCs while edited HSPCs are protected from CIM053-ADC in vitro.
- CD45 variant 3B edited HSPCs engraft and differentiate normally in vivo.
- A single dose of CIM053-ADC potently depletes unedited CD45 expressing cells while shielded hematopoietic cells persist.
- Collectively, our results demonstrate the identification of a molecular shield that can be installed in HSPCs by an adenine base editor. When paired with the CD45 targeting CIM053-ADC, it may enable a near universal strategy for the chemotherapy and irradiation-free replacement of the hematopoietic system. The combination therapy may also enable tumor-selective post-transplant maintenance therapy or applications beyond malignancies.

Acknowledgements

- Research Core Facilities (animal husbandry, flow cytometry) at the University of Basel and Department of Biomedicine.
- Funding from European Research Council (ERC) European Union's Horizon 2020 research and innovation programme (grant agreement No. 818806 (LTJ))
- Sponsored Research Collaboration Agreement (University of Basel/Cimeio Therapeutics AG (LTJ))

Contact

Jeker Lukas
lukas.jeker@unibas.ch

References

1. Palchaudhuri, R., Saez, B., Hoggatt, J. et al. Nat Biotechnol 2016
2. Walton RT. et al. Science 2020

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com